PE7.5/9

Quadruple Nucleoside Reverse Transcriptase Inhibitor (NRTI) Therapy with Trizivir (TZV) and Viread (TDF) in Naive and Experienced HIV/AIDS Patients: A 30-Month Follow-up

[1]Greiger-Zanlungo P, [2]Blick G, [3]Rubin D, [2]Gretz S, [2]Garton T, [2]Arzu Z, [1]Dupree D, [1]Gupta S, [1]Gonsalves G, [4]D Shepp, [4]R Stevens

[1]Mt Vernon Hosp/NY Medical College, Mt Vernon, NY; [2]Circle Medical LLC, Norwalk, CT; [3]New York Hosp Queens, Flushing, NY; [4]Gilead Sciences, Foster City, CA

10th European AIDS Conference/EACS, Dublin, Ireland, November 17-20, 2005

CONTACT INFO:
Gary Blick MD
153 East Ave,
Suite 32
Norwalk, CT 06851
BlickMD@CircleMedical.Org

BACKGROUND

1. Triple NRTI regimens may be class-sparing, but ACTG 5095 demonstrated TZV (ABC/3TC/AZT) alone inferior to EFV-containing regimens[1].
2. Use of once-daily, non-thymidine analog, triple NRTI regimens, e.g. ABC/3TC/TDF[2-4] and ddI/3TC/TDF[5], resulted in high rates of early virologic failure, possibly due to the low genetic barriers of the components used in these regimens.
3. Quadruple-NRTI regimens preserve other classes for future sequencing options, while lowering the risk of drug-drug interactions.
4. TDF is a potent and well-tolerated NtRTI and may improve the efficacy of TZV alone.
5. Several clinical trials have evaluated the use of TZV/TDF in treatment-naïve and -experienced individuals through 24-48 weeks:
 - Moyle et al.[6] compared TZV/TDF with CBV/EFV and showed equivalent viral load and CD4+ changes at week 48 in treatment-naïve individuals.
 - ESS300005 (ZIP)[7] demonstrated successful viral suppression through week 24 using TZV/TDF after first failure with AZT(or D4T)/3TC with a PI or nNRTI.
 - COL402638[8] evaluated TZV/TDF in ARV-naïve individuals and showed good virologic responses, especially with baseline PCR <100,000 c/ml, at week 24.
 - We previously demonstrated that TZV/TDF provided potent and durable viral suppression for >12 months in ARV-naïve and -experienced patients, whether intensifying with TDF or simplifying HAART[9].
6. We present the longest follow-up to-date on patients taking TZV/TDF with our original 75 patient cohort having been followed for up to 46 months (mean 30 months).

METHODS

- Prospective safety/efficacy follow-up of our 75 patients taking TZV/TDF from 3 different clinics:
 - Mt. Vernon Hospital HIV/ AIDS Clinic, Mt. Vernon, NY: inner city clinic
 - CIRCLE Medical LLC, Norwalk, CT: private practice setting
 - New York Hospital, Queens HIV/AIDS Clinic, Flushing, NY: private practice and public hospital clinic settings
- Patients were stratified into two GROUPS and the treatment-experienced group was stratified into two ARMS:
 - **Group A**: HAART-naïve (n=25)
 - **Group B**: HAART-experienced (n=50)
 - **Arm 1**: TZV intensification with TDF (n=21)
 - **Arm 2**: HAART simplification to TZV/TDF (n=29)
- Patients were selected for analysis as follows:
 - **Group A:** if they had taken at least 4 weeks of TZV/TDF
 - **Group B:**
 - **Arm 1**: if they were taking TZV alone, were **intensified** with TDF regardless of PCR, and took at least 4 weeks of TZV/TDF
 - **Arm 2**: if they **simplified** HAART, began TZV/TDF for any reason (choice, AE, rebound viremia, etc.), and took at least 4 weeks of TZV/TDF
- Virologic Failure was defined as:
 - Failure to attain a viral load <400 copies/ml by study end
 - Viral Rebound by ≥0.5 log (i.e. to ≥1265 copies/ml) after achieving a viral load <400 copies/ml
- Data was updated and analyzed through October, 2005 following submission of the abstract in July, 2005.

RESULTS

Demographics

TABLE 1: Baseline Demographics: Age, Gender, Race, Risk, Time HIV+, % AIDS, % HCV

	Group A (Naïve, n=25)	Group B (Experienced, n=50)
Age: median, yrs. (range)	42 (31-57)	42 (23-65)
Gender: Male, %	72	76
Race:		
Caucasian, n (%)	15 (60)	18 (36)
Black/AA, n (%)	8 (32)	18 (36)
Hispanic, n (%)	2 (8)	14 (28)
Risk Factor, HIV:		
MSM, n (%)	12 (48)	27 (54)
Heterosexual contact, n (%)	10 (40)	15 (30)
Injection drug use, n (%)	3 (12)	10 (20)
Other/Unknown, n (%)	0	1 (2)
Time HIV+: mean, mos. (range)	33 (18-59)	74 (14-168)
AIDS, n (%)	6 (24)	30 (60)
HIV/HCV, n (%)	5 (20)	10 (20)

In Group B, Arm 1 (intensify) had more Caucasian, MSM, males than Arm 2 (simplify), which had more Black/AA, heterosexual, females.

RESULTS (continued)

TABLE 2: Baseline Demographics: CD4, Viral Load at 1st HAART and TZV/TDF Start, ARV History

	Group A (Naïve, n=25)	Group B (Experienced, n=50) Arm 1: (intensify, n=21)	Group B (Experienced, n=50) Arm 2: (simplify, n=29)
Mean Prior ARVs, # (range)	N/A	4 (3-6)	6 (3-11)
Mean Time Prior ARVs, mos (range)	N/A	42 (19-80)	43 (7-101)
% Prior ARV (TZV, TDF) use			
AZT	N/A	100	59
3TC	N/A	100	90
ABC	N/A	100	41
TDF	N/A	0	14
1st HAART:			
Median CD4+, cells/mm3 (range)	N/A	207 (0-646)	218 (16-518)
Median CD4% (range)	N/A	15 (0-38)	13 (1-29)
Mean HIV-1 RNA, log_{10} (range)	N/A	5.1 (3.3-5.7)	5.3 (3-6)
TZV/TDF Start:			
Median CD4+, cells/mm3 (range)	340 (128-625)	471 (204-840)	435 (5-1334)
Median CD4% (range)	22 (4-35)	27 (13-45)	21 (1-37)
Mean HIV-1 RNA, log_{10} (range)	4.7 (4.2-5.1)	2.9 (1.7-5.2)	3.1 (1.7-5.6)
% <400 At Start	0	48	45

Adverse Events and Discontinuations

TABLE 3: Time on TZV/TDF, % Discontinuations, Serious Adverse Events

	Group A (Naïve, n=25)	Group B (Experienced, n=50) Arm 1: (intensify, n=21)	Group B (Experienced, n=50) Arm 2: (simplify, n=29)
Mean Time TZV/TDF, mos. (range)	33 (25-46)	27 (8-42)	29 (10-43)
Discontinuations, n (%)	3 (10)	4 (19)	6 (21)
Mean Time on TZV/TDF at discontinuation, mos. (range)	26 (25-27)	20 (11-32)	20 (10-31)
Reasons for discontinuation:			
Adverse event, n (events)	1 (GI)	3 (2 GI, 1 lactic acidosis/↑SCr)	0
Noncompliance, n	2	0	3
Virologic Failure, n (%)	0	1 (5)	2 (7)
Other, n (reason)	0	0	1 (STI)
Serious Adverse Events:			
%HSR	0	0	0
Grade III/IV SCr, n (%)	0	1 (5)	0
Grade III/IV AEs, n (%, type)	0	1 (5, lactic acidosis)	0

Immunologic and Virologic Responses

TABLE 4: Immunologic (median CD4) and Virologic (mean PCR) Responses

		BASELINE CD4%	BASELINE CD4#	BASELINE PCR (log_{10})	ON-TREATMENT CD4%	ON-TREATMENT CD4#	ON-TREATMENT PCR (log_{10})
Group A: (Naïve, n=25)		22 (4-35)	340 (128-625)	4.7 (4.2-5.1)	33 (12-51)	612 (287-1013)	2.1 (1.7-3.5)
Group B: (Experienced n=50)	Overall	25 (1-45)	398 (5-1334)	3.0 (1.7-5.6)	28 (1-44)	515 (7-1122)	2.2 (1.7-5.1)
	Arm 1: (intensify n=21)	27 (13-45)	471 (204-840)	2.9 (1.7-5.2)	30 (17-44)	599 (311-829)	2.0 (1.7-3.2)
	Arm 2: (simplify, n=29)	21 (1-37)	376 (5-1334)	3.1 (1.7-5.6)	26 (1-42)	507 (7-1122)	2.3 (1.7-5.1)

- Median % Change in CD4 cell count from Baseline:
 - Group A: +73%
 - Group B (Arm 1, Intensify): +27%
 - Group B (Arm 2, Simplify): +39%
- Median % Change in CD4% from Baseline:
 - Group A: +55%
 - Group B: (Arm 1, Intensify): +23%
 - Group B: (Arm 2, Simplify): +24%

FIGURE 1. Interval Median Change in Absolute CD4+ Cell Count from Baseline

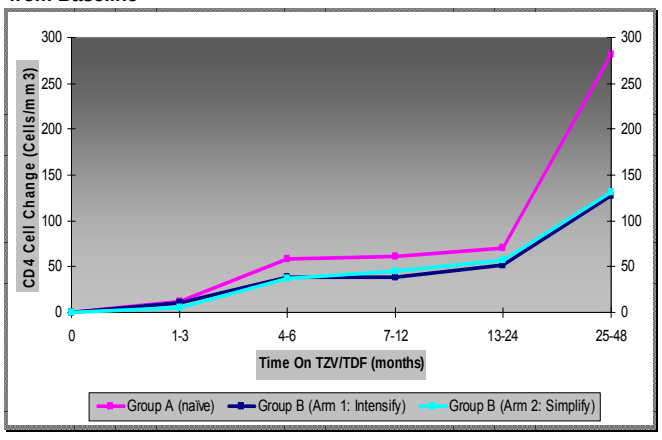
300
250
200
150
100
50
0
CD4 Cell Change (Cells/mm3)
0
1-3
4-6
7-12
13-24
25-48
Time On TZV/TDF (months)
Group A (naïve)
Group B (Arm 1: Intensify)
Group B (Arm 2: Simplify)

FIGURE 2. Interval Median Change in CD4% from Baseline

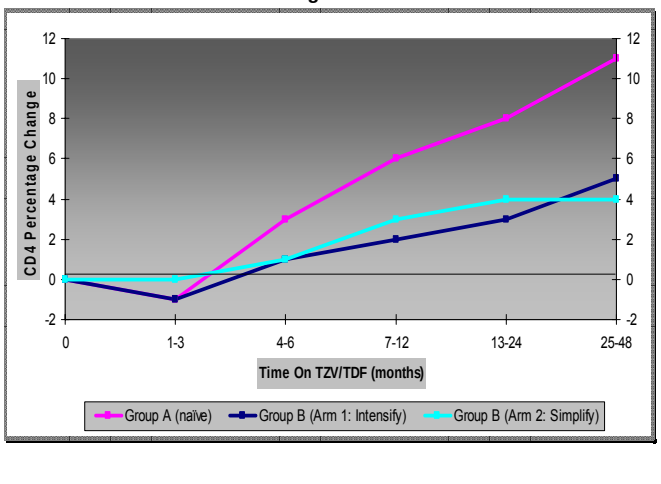
12
10
8
6
4
2
0
-2
CD4 Percentage Change
0
1-3
4-6
7-12
13-24
25-48
Time On TZV/TDF (months)
Group A (naïve)
Group B (Arm 1: Intensify)
Group B (Arm 2: Simplify)

RESULTS (continued)

TABLE 5. Virologic Responses
(*ITT, M/NC=F: intention-to-treat, missing/never completed=failure)

	Group A (Naïve, n=25)	Group B (Experienced, n=50) Arm 1: (intensify, n=21)	Group B (Experienced, n=50) Arm 2: (simplify, n=29)
PCR <400 copies/ml (%):			
At Baseline	0	48	45
On TZV/TDF, At least once	100	95	97
On TZV/TDF, Mo.12; ITT, M/NC=F*	88	76	86
On TZV/TDF, Study end; ITT, M/NC=F*	80	81	72
PCR <50 copies/ml (%):			
At Baseline	0	14	14
On TZV/TDF, At least once	92	76	72
On TZV/TDF, Mo.12; ITT, M/NC=F*	60	48	55
On TZV/TDF, Study end; ITT, M/NC=F*	60	62	48
PCR Change from Baseline:			
Baseline PCR, copies/ml (log_{10})	54,953 (4.7)	12,156 (2.9)	24,815 (3.1)
Mean copies/ml, log_{10}	-2.6	-0.8	-0.74
Virologic Failure, n (%):			
As Treated	12	5	17
ITT, M/NC=F*	12	19	21

Resistance Data

- One patient developed a K65R with virologic failure.
- Group A: Of the 3 patients meeting protocol-defined virologic failure, only 1 continued on HAART, was salvaged with ABC/TVD/ATZ/r without genotypic data, and resuppressed to PCR<400 within four months of the new regimen.
- Group B, Arm 1:
 - 1 pt. experienced virologic rebound (As Treated) with PCR 1421 and was successfully treated with ABC/TVD/FPV/r with genotypic mutations at 41, **K65R**, 67, 184, 210, and 215. The possibility of superinfection exists from patient history.
 - 3 patients experiencing AEs, 1 with PCR 825 and 2 with PCR <50, were successfully salvaged with ABC/3TC/ATZ/r, TVD/EFV, and ddI/TVD/ATZ/r, respectively.
- Group B, Arm 2:
 - Of 5 pts. with virologic failure, 1 remains on TZV/TDF, 1 is lost-to-follow-up, and 3 were resuppressed to PCR<400 after a mean 4.5 months with:
 - TVD/ATZ/r (PCR 1100, no GT),
 - ddI/TVD/ ATZ/r (PCR log_{10} 5.13, GT: RT: 41,67,74,103,118,184,210,215; PI: 54,63,71,82),
 - ABC/TVD/ ATZ/r (PCR 3280, GT: 41, 67,184,210).

CONCLUSIONS

1. The quadruple-NRTI regimen of TZV/TDF safely provides adequate and durable viral suppression without evidence of significant virological failure for up to 46 months with impressive immunological improvements in both HAART-naïve and HAART-experienced patients, whether intensifying TZV with TDF or simplifying HAART with TZV/TDF.
2. The improved outcome seen in this study compared to that seen with triple NRTI combinations, such as AZT/ABC/3TC[1] and ABC/TDF/3TC[2-4], suggests that ABC and TDF can be combined effectively as part of an appropriately configured regimen.
3. The improved outcome may be attributable to the alternate pathway to resistance imposed by inclusion of AZT (i.e., TAM-mediated nucleotide excision) and the relative incompatibility of the K65R pathway, selected for by both ABC and TDF, with the TAM pathway.
4. Future studies are needed to determine if this quadruple-NRTI approach is superior to TZV alone or comparable to multi-class regimens employing an NRTI backbone consisting of only 2 of the 4 NRTIs (ABC/3TC, TDF/3TC or FTC) used in this study.

REFERENCES

1. Gulick RM, Ribaudo HJ, Shikuma CM, et al, for the AIDS Clinical Trials Group Study A5095 Team. Triple-nucleoside regimens versus efavirenz-containing regimens for the initial treatment of HIV infections. NEJM. 2004;350:1850-1861.
2. Gallant JE, Rodriguez AE, Weinberg W. Early non-response to tenofovir DF (TDF) + abacavir (ABC) and lamivudine (3TC) in a randomized trial compared to efavirenz (EFV) + ABC and 3TC: ESS30009 unplanned interim analysis. Program and Abstracts of the 43rd Interscience Conference on Antimicrobial Agents and Chemotherapy; September 14-17, 2003; Chicago, Illinois. Oral presentation H-1722a.
3. Farthing C, Khanlou H, Yeh V. Early virologic failure in a pilot study evaluating the efficacy of once daily abacavir (ABC), lamivudine (3TC) and tenofovir DF (TDF) in treatment naive HIV-infected patients. Program and Abstracts of the 2nd International AIDS Society Conference on Pathogenesis and Treatment; July 13-16, 2003; Paris, France. Abstract 43.
4. Landman R, Peytavin G, Descamps D, et al. Low genetic barrier to resistance is a possible cause of early virologic failures on once-daily regimen of abacavir, lamivudine, and tenofovir: The TONUS Study. Program and Abstracts of the 11th Conference on Retroviruses and Opportunistic Infections; February 8-11, 2004. San Francisco, California. Abstract 52.
5. Jemsek J, Hutcherson P, Harper E. Poor virologic responses and early emergence of resistance in treatment naive, HIV-infected patients receiving a once daily triple nucleoside regimen of didanosine, lamivudine and tenofovir DF. Program and Abstracts of the 11th Conference on Retroviruses and Opportunistic Infections; February 8-11, 2004; San Francisco, California. Abstract 51.
6. Moyle G, Nelson M, Higgs C, Mandalla S, Fisher M, Johnson M, Gazzard B. A randomized open label comparative study of Combivir + efavirenz (2 class triple therapy) versus Trizivir + tenofovir (single class quadruple therapy) in initial therapy for HIV-1 infection. Program and Abstracts of the 44th Interscience Conference on Antimicrobial Agents and Chemotherapy; October 30-November 2, 2004, Washington, DC. Abstract H-1131.
7. Rodriguez AE, Hill-Zabala CE, Sloan LA, Jefferson TT, Yau L, Johnson J, Shaeffer MS. Abacavir/Lamivudine/Zidovudine (TZV) + Tenofovir (TDF) in Subjects With Early Virologic Failure on an Initial Regimen of Zidovudine (ZDV) or Stavudine (d4T) + Lamivudine (3TC) and a Protease Inhibitor (PI) or Non-Nucleoside Reverse Transcriptase Inhibitor (NNRTI) (ESS30005, ZIP). Program and Abstracts of the 44th Interscience Conference on Antimicrobial Agents and Chemotherapy; October 30 - November 2, 2004; Washington, DC. Abstract H-563.
8. DeJesus E, Elion R, Cohen C, Redfield R, Gathe J, Hsu R, Yau L, Ross L, Ha B. Week 24 Analysis of Once-Daily Trizivir (TZV) and Tenofovir DF (TDF) in Antiretroviral-Naive Subjects (COL40263). Program and Abstracts of the 44th Interscience Conference on Antimicrobial Agents and Chemotherapy; October 30 - November 2, 2004; Washington, DC. Abstract H-564.
9. Greiger-Zanlungo P, Rubin D, Blick G, Garton T, Arzu Z, Shepp D. Quadruple Nucleoside Reverse Transcriptase Inhibitor (NRTI) Therapy with Trizivir (TZV) and Viread (TDF) in HAART-Naïve and HAART–Experienced HIV/AIDS Patients. Program and Abstracts of the XV International AIDS Conference, Bangkok, Thailand, July 11-16, 2004. Abstract TuPeB4513.